Population pharmacokinetics of belimumab in systemic lupus erythematosus: insights for monoclonal antibody covariate modeling from a large data set

Herbert Struemper (1), Wendy Cai (2)

(1) Clinical Pharmacology Modeling & Simulation, GlaxoSmithKline, RTP, NC & King of Prussia, PA, USA; (2) Human Genome Sciences, Rockville, MD, USA.

Introduction

- Belimumab is a recombinant, human immunoglobulin (Ig)-G1λ monoclonal antibody (mAb) that targets B-lymphocyte stimulator (BLyS)
- BLyS is a cytokine that promotes B-cell selection, survival, and differentiation [1]
- Belimumab is indicated for the treatment of adult patients with active, autoantibody-positive systemic lupus erythematosus (SLE) receiving standard therapy
- Belimumab is also under investigation in lupus nephritis, membranous nephropathy, antineutrophil cytoplasmic antibodies-associated vasculitides, chronic graft-vs-host disease, myastenia gravis and other auto-immune related conditions.

Objectives

- Characterize the population pharmacokinetics of belimumab following intravenous infusion in patients with SLE
- Identify factors contributing to the pharmacokinetic variability

Methods

Data

- PK data from one 1 Phase I, 1 Phase II, and 2 Phase 3 studies with a total of 1603 patients and 8143 observations:
 - Phase 1: single dose or 2 doses 28 days apart; 1, 4, 10, and 20 mg/kg; N=57 (Clinical Trials.gov registry number: NCT00657007)
 - Phase 2: Multiple doses on d 0, 14, and 28, and then every 28 d; 1, 4, and 10 mg/kg; N=424 (NCT00071487)
 - Phase 3 (BLISS-52): Multiple doses on d 0, 14, and 28, and then every 28 d through 48 wk, 1 and 10 mg/kg, N=578 (NCT00424476)
 - Phase 3 (BLISS-76): Multiple doses on d 0, 14, and 28, and then every 28 d through 72 wk, 1 and 10 mg/kg, N=544 ([NCT00410384)

PK Modeling

- Non-linear mixed effects modeling approach using NONMEM® (ADVAN6): Models were prioritized using objective function values, mechanistic plausibility of parameters, and performance in visual predictive checks.
- Stepwise covariate model building assumed a 0.001 α-level significance threshold for forward addition and backward elimination.
- Main model developed with categorical proteinuria covariate (< or ≥ 2 g/day); subsequent modeling included continuous proteinuria covariate.

Results

- Exposure was approximately dose proportional and well characterized by a linear two-compartment PK model with CL from central compartment with a terminal half-life of 19 days, Vss of 5.3 L and CL of 0.22 L/day for the 10 mg/kg dose in the Phase 3 trials (Fig. 1).
- 16 covariate effects of which 9 were related to patient characteristics were identified.
- Baseline BlyS level was not identified as a statistically significant covariate confirming other evidence that target mediated disposition did not have a substantial effect for tested dose levels (see [2] for more detail).

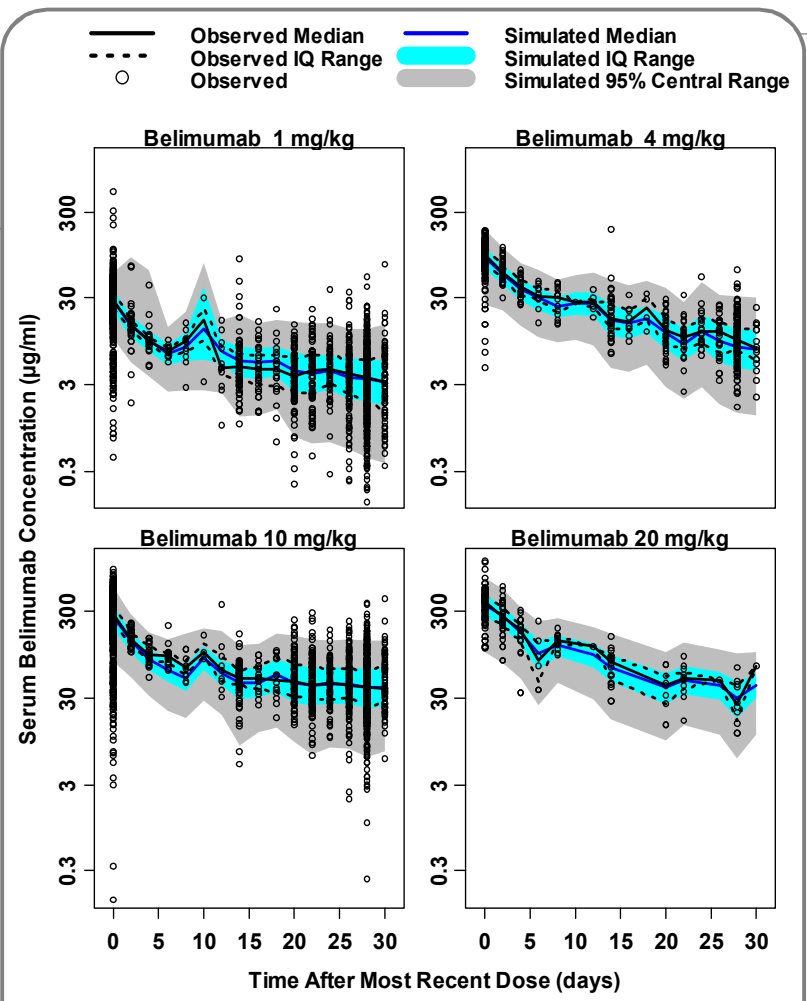
Observed Median
Observed IQ Range
Observed
Simulated Median
Simulated IQ Range
Simulated 95% Central Range
Belimumab 1 mg/kg
Belimumab 4 mg/kg
Belimumab 10 mg/kg
Belimumab 20 mg/kg
Serum Belimumab Concentration (µg/ml)
0.3
3
30
300
0 5 10 15 20 25 30
Time After Most Recent Dose (days)

Fig. 1: VPC, observed vs simulated belimumab conc.

Model Parameters

Parameter	Implementation	Model Estimate (%RSE)	BST Estimate (%RSE)
Fixed effects			
CL, mL/d	THETA (1)	215 (1.95)	215 (1.95)
Effect of BWT	x $(BWT/66.3)^{THETA\ (5)}$	0.506 (11.1)	0.507 (6.47)
Effect of BCCL	x $(BCCL/79.9)^{THETA\ (8)}$	0.217 (11.3)	0.214 (11.4)
Effect of SPUS	x (1+ THETA [9])	0.143 (35.2)	0.144 (34.7)
Effect of ACEI	x (1+ THETA [11])	0.0851 (21.4)	0.0842 (22.1)
Effect of Phase 1 study	x (1+ THETA [12])	0.949 (6.90)	0.953 (6.94)
Effect of Phase 2 study	x (1+ THETA [13])	0.717 (4.10)	0.721 (4.03)
Effect of STER	x (1+ THETA [17])	0.0603 (22.7)	0.0604 (21.2)
Effect of BALB	x $(BALB/4.0)^{THETA\ (19)}$	-0.829 (9.49)	-0.834 (-9.71)
Effect of BIGG	x $(BIGG/1480)^{THETA\ (20)}$	0.322 (6.96)	0.324 (7.38)
V1, mL	THETA (2)	2560 (0.961)	2560 (1.00)
Effect of BWT	x $(BWT/66.3)^{THETA\ (6)}$	1.14 (7.38)	1.15 (7.20)
Effect of BBMI	x $(BBMI/25.1)^{THETA\ (7)}$	-0.616 (15.7)	-0.623 (-14.5)
Effect of BHGB	x $(BHGB/12.4)^{THETA\ (10)}$	-0.291 (23.3)	-0.281 (-24.8)
Effect of Phase 1 study	x (1+ THETA [14])	0.366 (12.7)	0.367 (12.4)
Effect of Phase 2 study	x (1+ THETA [15])	0.269 (6.69)	0.270 (6.42)
Effect of BWBC	x $(BWBC/5.6)^{THETA\ (18)}$	-0.0810 (20.0)	-0.082 (-19.6)
Q, mL/d	THETA (3)	459 (10.8)	464 (12.3)
V2, mL	THETA (4)	2730 (4.03)	2740 (5.14)
Effect of RDOS	x $(RDOS/10)^{THETA\ (16)}$	0.379 (8.34)	0.374 (8.43)
Interindividual variability			
ω^2_{CL}	OMEGA (1,1)	0.0699 (6.27)	0.0696 (6.64)
ω^2_{V1}	OMEGA (2,2)	0.0398 (28.4)	0.0377 (33.1)
$\omega^2_{CL/V1}$	OMEGA (2,1)	0.0261 (17.3)	0.0258 (17.3)
ω^2_{V2}	OMEGA (3,3)	0.0969 (35.2)	0.0960 (37.3)
Residual variability			
$\sigma^2_{proportional}$	SIGMA (1)	0.0887 (4.01)	0.0887 (3.84)
$\sigma^2_{additive}$	SIGMA (2)	0.0139 (32.8)	0.0137 (37.1)

%RSE, percentage relative standard error, calculated as (SE ÷ mean)*100%; BST Estimate, bootstrap estimate based on 1029 successful bootstrap runs (86% of 1200 runs minimized successfully); THETA, structural model parameter in NONMEM code; CL, systemic clearance; V1, central volume of distribution; Q, intercompartmental clearance, V2, peripheral volume of distribution; BWT, baseline body weight; BCCL, baseline calculated creatinine clearance; SPUS, baseline proteinuria level group; ACEI, baseline angiotensin-converting enzyme inhibitor use; STER, corticosteroid use; BALB, baseline albumin level; BIGG, baseline immunoglobulin-G level; BBMI, baseline body mass index; BHGB, baseline hemoglobin; BWBC, baseline white blood cell count; RDOS, dose group.

IgG effect on CL and $T_{1/2}$

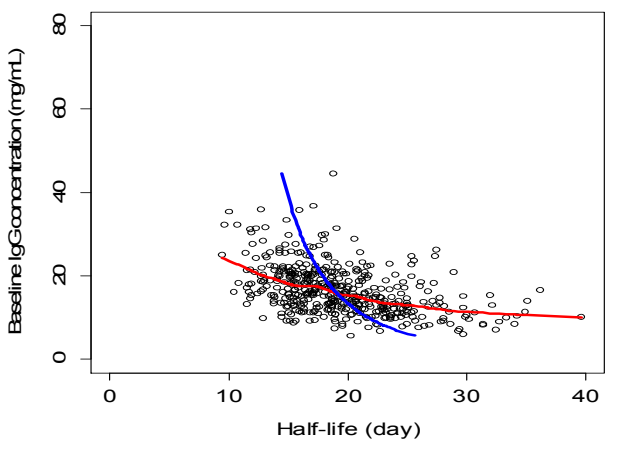
Baseline IgG concentration (mg/mL)
0
20
40
60
80
0 10 20 30 40
Half-life (day)

Fig. 2: Baseline IgG concentration vs half-life (post-hoc parameter estimates) in Phase 3 subjects with I< 0.5 g/day proteinuria receiving 10 m/kg belimumab (red curve, Loess fit; blue curve; half-life based on population parameters and estimated IgG effect; compare to [4] Fig. 8)

Proteinuria effect on CL

- After proteinuria status (< or ≥2 g/day) was identified as a significant covariate, the effect of proteinuria as a continuous variable was analyzed in separate model
- A *linear* relationship best described proteinuria effect on the typical value of CL (TVCL) among functional relationships tested (Fig. 3) :
 - TVCL = ($TVCL_{w/o\ proteinuria\ effect}$ + 18.7 mL/g * proteinuria
- Effect on CL relatively minor in tested SLE population
- Larger effect size predicted for patient populations with larger levels of proteinuria (membranous nephropathy, etc.), e.g.:
 - 10 g/d proteinuria → 89 % increase in CL → 47% decrease in AUC relative to belimumab CL with no/negligible proteinuria

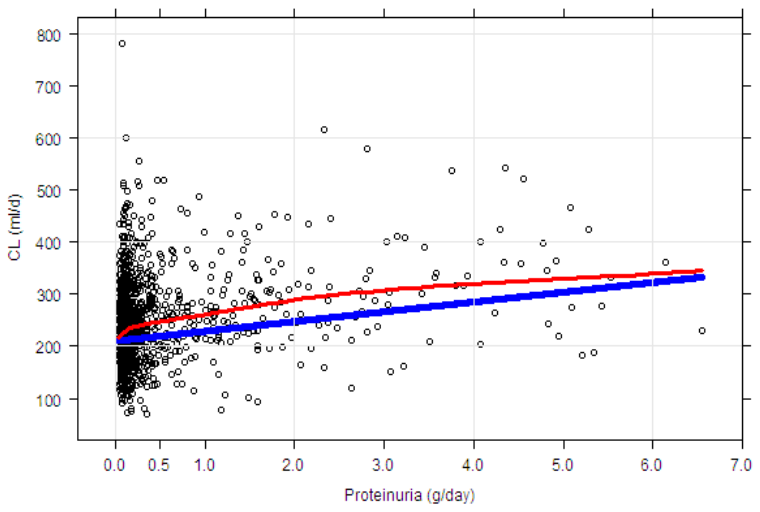
CL (ml/d)
100
200
300
400
500
600
700
800
0.0 0.5 1.0 2.0 3.0 4.0 5.0 6.0 7.0
Proteinuria (g/day)

Fig. 3: Baseline proteinuria vs CL (post-hoc parameter estimates) in Phase 3 subjects for model with continuous proteinuria effect (red curve, Loess fit; blue line; effect based on population parameters)

Conclusions

- Belimumab PK in SLE was well characterized by a linear 2-compartment PK model; PK parameters were consistent with other IgG1 mAbs [3].
- Effect of proteinuria on CL was identified in SLE population, which might be clinically relevant for disease populations with higher levels of proteinuria. Further evaluation is under way in membranous nephropathy.
- Effect of IgG on CL was detectable in the SLE population (hypergammagobulinemia trend) and consistent with hypothesis and data for saturation of the neonatal Fc receptor (FcRn) recycling at high endogenous IgG levels [4].
- Effect of baseline albumin (BALB) on CL not inconsistent with IgG effect as albumin binds to a different epitope of FcRn and therefore does not compete with IgG for FcRN binding [4].
- Effect of BHGB on V1 was detectable and qualitatively consistent with decreased serum volume due to increased hematocrit (estimated effect was approx. half the effect predicted with assumption of hematocrit [%] ~ 3x BHGB [g/dL]).
- None of the covariate effects were found to require dose adjustment in the SLE population.

References

[1] Cancro MP, D'Cruz DP, Khamashta MA. The role of B lymphocyte stimulator (BLyS) in systemic lupus erythematosus. J Clin Invest. 2009;119:1066-1073.
[2] Struemper H, Chen C, Cai W. Population Pharmacokinetics of Belimumab Following Intravenous Administration in Patients With Systemic Lupus Erythematosus. J Clin Pharm. 2013; published online 16 May 2013; DOI: 10.1002/jcph.104.
[3] Dirks NL, Meibohm B. Population pharmacokinetics of therapeutic monoclonal antibodies. Clin Pharmacokinet. 2010;49:633-659.
[4] Waldmann TA, Strober W. Metabolism of immunoglobulins. Prog Allergy. 1969;13:1–110.

Acknowledgments

At the time this work was initiated Herbert Struemper was employed by GlaxoSmithKline and Wendy Cai was employed by Human Genome Sciences. The authors wish to thank Cecil Chen, Thi-Sau Migone, Kevin Koch, Roxanne Jewell, and Peter Bonate.

Web Broadcasting Service
Created by live | video | interactive | design